AICOG 2025
Mumbai
67th All India Congress of Obstetrics & Gynaecology
FOGSI celebrates 75 years - Diamond Jubliee
8-12 January, 2025 | JIO World Convention Centre

Abstract Number: **EP191** | Name: **Dr. Pragya Pandey**

Effectiveness and Safety of Isoxsuprine Hydrochloride as Tocolytic Agent in Women with Preterm Labor in India - A Prospective, Observational Study

Shrinivas N Gadappa[1], Mary Snigdha[2], Arun Nayak[3], Pragya Pandey[4]

1. Government Medical College, Aurangabad, Maharashtra 2. AC Subbareddy Govt Medical College and hospital , Nellore, Andhra Pradesh 3. LTM Medical College, Sion, Mumbai, Maharashtra 4. Shubam Sadbhavna hospital, Varanasi, Uttar Pradesh

Introduction & Objectives

Preterm labor (PTL) is defined as labor occurring between 20 to 37 gestation weeks. Amongst the developing countries, India has a very **high incidence of PTL (23.3%).** PTL management utilizes tocolytic therapy to extend gestation which allows usage of corticosteroids and MgSO4.

Although β-agonists are extensively used for tocolysis, various studies have reported undesirable safety profiles. Among β-agonists, Isoxsuprine is being used in India for last six decades and **approved by CDSCO** for the management of PTL. Although, **Isoxsuprine is known to be effective, safe and well tolerated tocolytic agent,** there is a lack of recent evidence supporting efficacy and safety of molecule. The current study re-assessed the effectiveness and safety of isoxsuprine in managment of preterm labor in an Indian Multicenter setting.

Materials & Methods

N=170 Women with diagnosis of PTL were treated at onset of PTL with an **intravenous infusion of Isoxsuprine hydrochloride followed by oral isoxsuprine therapy** (40 mg bd- daily) Assessment done at **12, 24 & 48 hours** of initiation of therapy & telephonic follow-up for delivery outcome.

4 investigators from 4 centres across India participated in study.

1. Dr. Shrinivas N Gadappa (Aurangabad)
2. Dr Mary Snigdha (Nellore)
3. Dr Arun Nayak (Mumbai)
4. Dr Pragya Pandey (Varanasi)

Primary Endpoint: Percentage of women achieving Successful tocolysis at 24 and 48 hours after start of therapy

Secondary Endpoints: Percentage of women with (1) ADRs, (2) ADRs leading to discontinuation of isoxsuprine (3) Failure of tocolysis (4) Reduction in number of contractions at 12 hours

Exploratory Endpoints: (1) Percentage of women with healthy babies (2) Mean Birth Weight

Results & Discussion

■ Successful tocolysis (165)
■ Tocolysis failure (5)

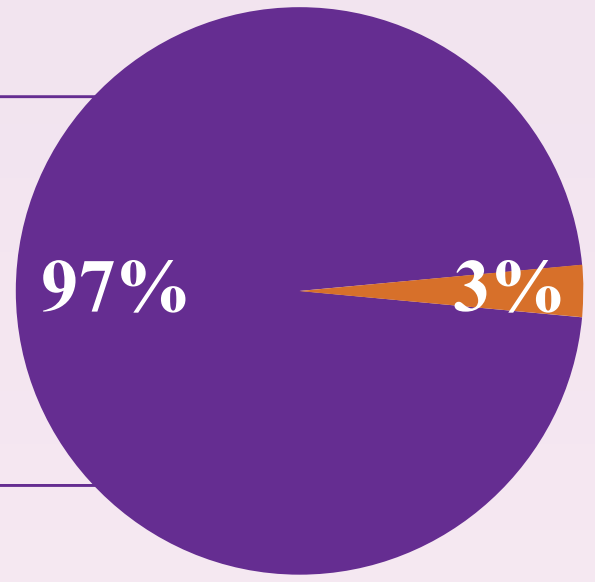
97%
3%

99 patients (58%) achieved Reduction in uterine contractions after 12 hours of therapy

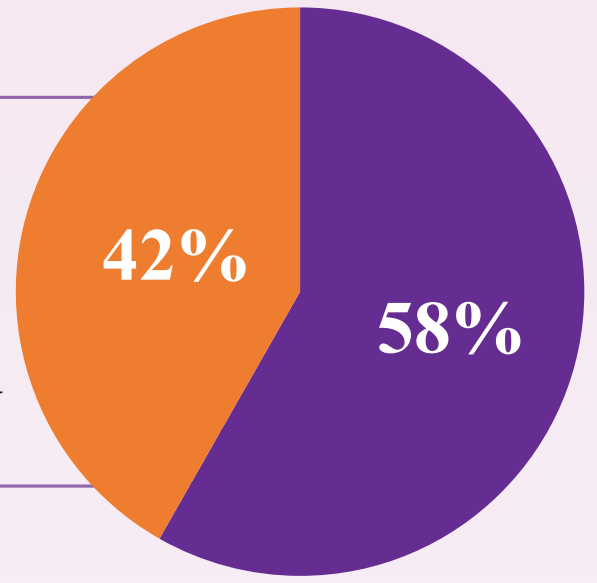
42%
58%

Perinatal Outcomes

Mean Birth weight= 2.7 kg

Healthy babies =146(85.9%)

Successful tocolysis rate (97.1%) indicate that isoxsuprine provided sufficient time for corticosteroids minimizing incidence of neonatal complications.

ADR= 10 (5.9%)

Lack of efficacy: 5 (2.9%)	Still birth: 2(1.2%)
Fetal Distress: 2 (1.2%)	Scar pain: 1(0.6%)

In response to AEs, **no action was taken with use of Isoxsuprine therapy. No clinically significant abnormality** like tachycardia or hypotension were reported

Isoxsuprine when used **in management of preterm labor resulted** in favorable outcomes in terms of birth weight and healthy babies (no congenital abnormalities)

Conclusion: Isoxsuprine was found to be an effective & well tolerated tocolytic agent in patients at risk of preterm delievery resulting in favorable maternal & perinatal outcomes

Acknowledgement: The authors thank the study participants without whom this study would not have been accomplished.
1. Conflict of Interest: There is no conflict of interest declared by the authors.
2. Source of Funding: The research is funded by Abbott India Ltd.

Abbreviations: PTL: Preterm Labor; **ADR:** Adverse Drug Reaction; **AE:** Adverse Event

Reference: Data on File (Based on clinical study report) Effectiveness and Safety of Isoxsuprine Hydrochloride as Tocolytic Agent in Women with Preterm Labor in India—A Prospective, Open-Label Study.